A subject discontinuation phase consisted of safety procedures performed 24 hours after each subject's last dose.

The Effect of Different C_{max}/T_{max} Ratios on Euphoria and Liking Following Oral Oxycodone Dosing in Opioid-Experienced, Non-Dependent, Recreational Drug Users

Lynn R Webster MD[1], Brad Bath PhD[1], Robert A Medve MD[1], Kevin Jessing MA[1], Alan L. Mueller PhD[1], Alice Jackson RN[1]

[1] Lifetree Clinical Research and Pain Clinic

This was an investigator sponsored study funded by a grant from King Pharmaceuticals Inc.

Introduction

► Prescription opioid abuse is a serious problem that has attracted much attention in recent years. Particularly alarming are the high rates of non-medical use of prescription pain relievers among adolescents. In 2007, 9.6% of 12th graders reported nonmedical use of Vicodin® hydrocodone/ acetaminophen [APAP]) and 5.2% reported non-medical use of OxyContin® (oxycodone).

Despite the prevalence of oxycodone abuse, there is little information in the public domain regarding its human abuse liability. Of the published studies, none have evaluated a controlled-release formulation of oxycodone and none have reported on the pharmacokinetics (PK) of oxycodone or the relationship between its PK and abuse liability. Based on the limited information available on the abuse liability of oxycodone, further studies (particularly PK-PD) are needed to understand the problem occurring today with prescription opioid abuse.

The pharmaceutical industry is developing new formulations intended to be more difficult to manipulate and abuse. The putative benefit of the new formulations is based upon the PK and PD effects, yet there is little data to support the concept that different PK's will influence the abuse potential of a drug. Therefore, this study was designed to explore the relationship between abuse potential (determined by "likeability" and "euphoria") and the PK profile (Cmax) and (Tmax) over a clinically relevant range of doses of oxycodone.

Objective

► The primary objective of the study was to evaluate the relationship between the pharmacokinetics of oral oxycodone and the pharmacodynamic endpoints, which are specific measures of abuse-liability in opioid-experienced, non-dependent, recreational drug users.

Methods

► The randomized, double-blind, placebo-controlled, crossover study contained 2 cohorts of opioid-experienced, non-dependent, recreational drug users. All subjects underwent a naloxone challenge and were screened for the ability to discriminate between 40 mg oxycodone and placebo. Qualifying subjects proceeded to the double-blind abuse-liability phase. Inclusion criteria included18 to 50 years of age, male with a history of recreationally abusing opioids defined as having experienced a "high" on at least 5 occasions in the last 12 months and once in the past 90 days. Subjects who abused multiple drugs had to prefer opioids. They could not be physically dependent on any drug of abuse and/or alcohol (excluding nicotine), as assessed by the Investigator and DSM-IV criteria.

Methods

Cohort 1 explored the PK-PD relationship of oxycodone formulations. Cohort 2 explored the PK-PD relationship of different oxycodone doses.

►Cohort 1 received 1 treatment per day on 5 consecutive days: 40 mg IR oxycodone (Immediate Release) in a capsule (two 20 mg tablets); 40 mg CR oxycodone (Controled Release) in a capsule (1 tablet crushed); 40 mg CR oxycodone in a capsule (1 tablet intact); 80 mg CR oxycodone in a capsule (2 tablets intact); and placebo capsule.

► Cohort 2 received 1 treatment per day on 4 consecutive days: 20 mg IR oxycodone in a capsule (1 tablet); 40 mg IR oxycodone in a capsule (two 20 mg tablets); 80 mg IR oxycodone in a capsule (four 20 mg tablets); and placebo capsule.

The primary endpoints were the Drug Effects Questionnaire (DEQ) questions, "Do you like the drug?" (DEQ #4) and "How high are you now?" (DEQ #5).

PK endpoints included the Tmax, Cmax, and AUC at 0.25, 0.50, 0.75, 1, 1.5, 2, 2.5, 3, 3.5, 4, 4.5, 5, 6, 8, and 12 hours after dosing, calculated via plasma samples and compared to PD effects.

The relations between DEQ #4 and DEQ #5 scores at Tmax and Cmax were assessed. The relation of the log of the abuse quotient (AQ) to the AUE of DEQ #4 and DEQ #5 was assessed for intervals 0-15, 0-30, 0-45, 0-60, 0-90 minutes for each dose. The AQ is Cmax/Tmax.

Results

36 subjects were enrolled

18 subjects completed the abuse liability phase in cohort 1.
16 subjects completed the abuse liability phase in cohort 2.
No subject withdrew due to AE's or safety issues

Figures 1, Cohort 1 shows that the relation of Emax to Tmax could not be clearly assessed since Cmax was not held *as* constant as the study was attempting to perform by the original design; Cohort 2 shows excellent correlation of Emax to a change in Cmax for both DEQ 4 (R2 = 0.8448) and DEQ 5 (R2 = 0.9661).

Figure 2 shows the PK of all doses in Cohort 1 and 2

Figures 3 and 4 show the mean DEQ #4 and DEQ #5 at each PK sample time for both Cohort 1 and 2 .

Figure 5 shows excellent correlation of AQ to liking (DEQ #4) from 15 to 60 minutes and very good correlation up to 90 minutes using the AUE of DEQ #4 for each time point.

Figure 6 shows excellent correlation of AQ to euphoria (DEQ #5) up to 60 minutes but a lesser correlation from 0 to 90 minutes using the AUE of DEQ #5 for each time point

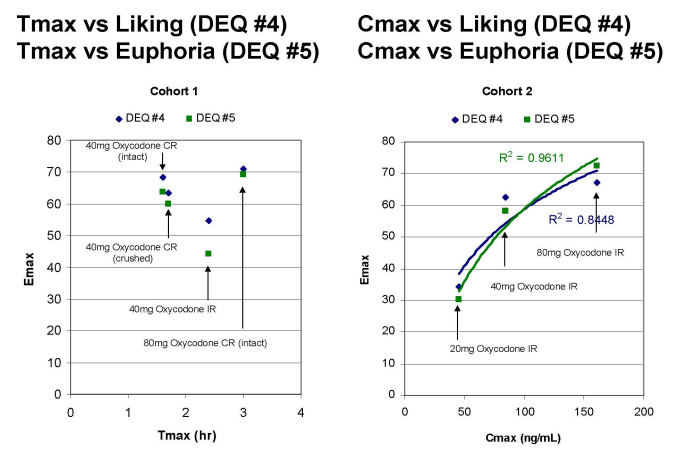

Tmax vs Liking (DEQ #4)
Tmax vs Euphoria (DEQ #5)
Cmax vs Liking (DEQ #4)
Cmax vs Euphoria (DEQ #5)
Cohort 1
Cohort 2
DEQ #4
DEQ #5
40mg Oxycodone CR (intact)
40mg Oxycodone CR (crushed)
40mg Oxycodone IR
80mg Oxycodone CR (intact)
80mg Oxycodone IR
20mg Oxycodone IR
R^2 = 0.9611
R^2 = 0.8448
Emax
Tmax (hr)
Cmax (ng/mL)

Figure 1

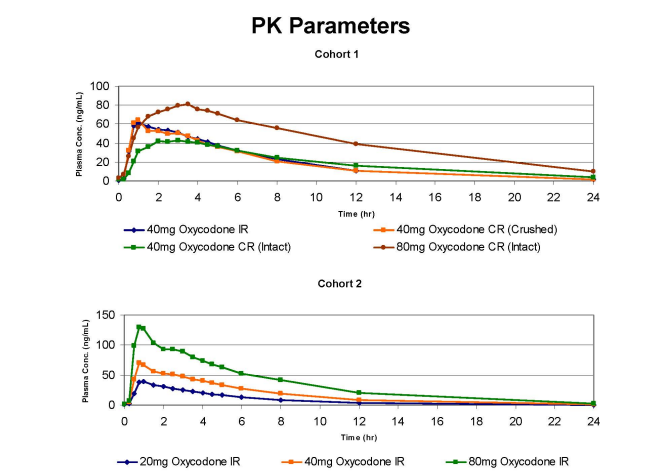

PK Parameters
Cohort 1
Plasma Conc. (ng/mL)
Time (hr)
40mg Oxycodone IR
40mg Oxycodone CR (Intact)
40mg Oxycodone CR (Crushed)
80mg Oxycodone CR (Intact)
Cohort 2
20mg Oxycodone IR
40mg Oxycodone IR
80mg Oxycodone IR

Figure 2

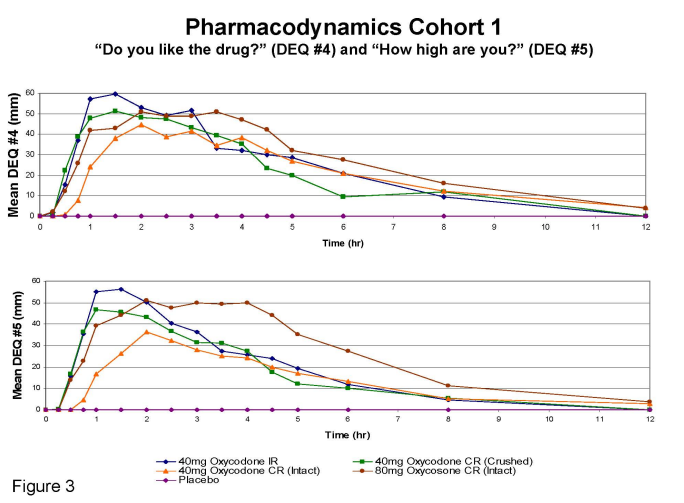

Pharmacodynamics Cohort 1
"Do you like the drug?" (DEQ #4) and "How high are you?" (DEQ #5)
Mean DEQ #4 (mm)
Mean DEQ #5 (mm)
Time (hr)
40mg Oxycodone IR
40mg Oxycodone CR (Intact)
Placebo
40mg Oxycodone CR (Crushed)
80mg Oxycosone CR (Intact)

Figure 3

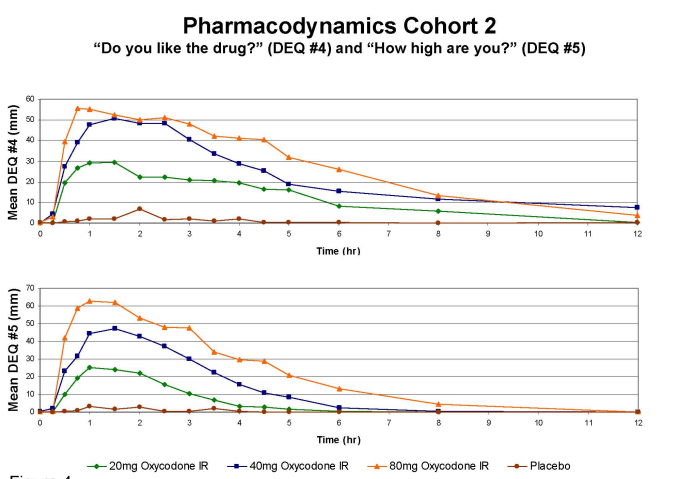

Pharmacodynamics Cohort 2
"Do you like the drug?" (DEQ #4) and "How high are you?" (DEQ #5)
Mean DEQ #4 (mm)
Mean DEQ #5 (mm)
Time (hr)
20mg Oxycodone IR
40mg Oxycodone IR
80mg Oxycodone IR
Placebo

Figure 4

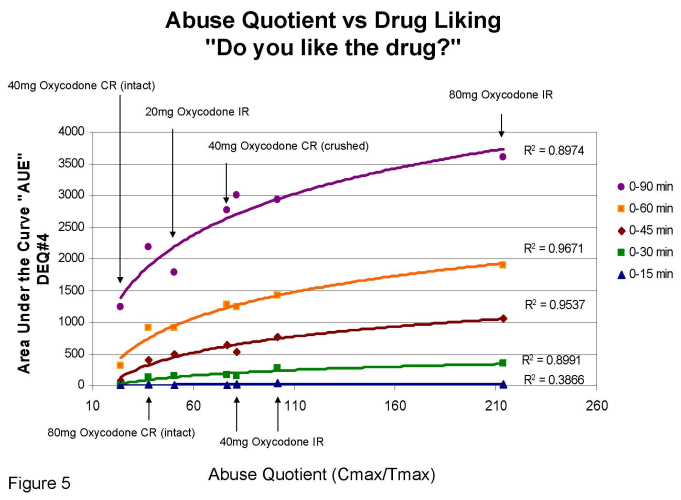

Abuse Quotient vs Drug Liking
"Do you like the drug?"
40mg Oxycodone CR (intact)
20mg Oxycodone IR
40mg Oxycodone CR (crushed)
80mg Oxycodone IR
80mg Oxycodone CR (intact)
40mg Oxycodone IR
R^2 = 0.8974
R^2 = 0.9671
R^2 = 0.9537
R^2 = 0.8991
R^2 = 0.3866
0-90 min
0-60 min
0-45 min
0-30 min
0-15 min
Area Under the Curve "AUE" DEQ#4
Abuse Quotient (Cmax/Tmax)

Figure 5

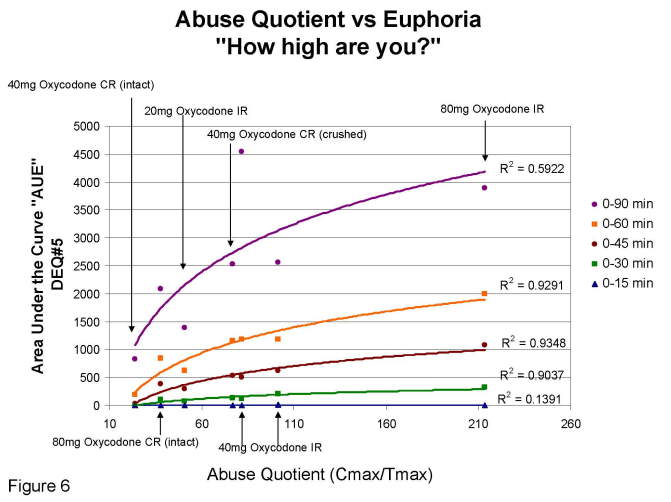

Abuse Quotient vs Euphoria
"How high are you?"
40mg Oxycodone CR (intact)
20mg Oxycodone IR
40mg Oxycodone CR (crushed)
80mg Oxycodone IR
80mg Oxycodone CR (intact)
40mg Oxycodone IR
R^2 = 0.5922
R^2 = 0.9291
R^2 = 0.9348
R^2 = 0.9037
R^2 = 0.1391
0-90 min
0-60 min
0-45 min
0-30 min
0-15 min
Area Under the Curve "AUE" DEQ#5
Abuse Quotient (Cmax/Tmax)

Figure 6

Discussion

This is the first study to show the relationship between oxycodone *liking and Euphoria to the pharmacokinetics, Tmax* and *Cmax* over a clinically relevant range of doses. Basic science research has suggested that the abuse potential of an opioid is related to its Cmax and Tmax but the relative importance of each has not been elucidated. This study suggest Cmax correlates well with the degree of " likeability" and magnitude of a "high". However we could not determine if a change in Tmax also correlates with Emax since the PK of Cmax across treatment groups was not controlled. Further exploration of this relationship is required. There was a strong correlation with the ratio of Cmax/Tmax which supports the construct that the evaluation of the AQ may be a reasonable assessment for the potential of abuse. This is due to the strong relationship found for the AQ ratio to the likeability of oxycodone (0 to 90 minutes). Likewise, excellent correlation was found for the AQ to euphoria (0 to 60 minutes). Increasing the Cmax (dose) during the 0-90 minutes did not produce proportional increase in euphoria. This could mean that once you are "high" additional drug can not continue to produce proportional euphoria. The PK data suggest whole CR oxycodone has an immediate release component which may influence the AQ,. Although this data does not support direct correlation of Tmax and Emax the AQ data suggest that this may have an influence on abuse potential, the extent of this influence would need to be further explored in future studies.

Conclusion

•A range of 20 to 80 mg IR oxycodone can be safely administered to healthy subjects for research purposes.

•Emax for "liking" and "euphoria" strongly correlates with Cmax

•If liking and euphoria effects drive oxycodone abuse, then an AQ value may help determine the abuse potential of oxycodone formulations. A high AQ value may help identity preferred drugs and formulations sought by recreational prescription drug abusers. Conversely a low AQ value may suggest less attractiveness as a drug of abuse.

• Further research is needed to determine if indeed this construct will hold true over time and with other opioids and classes of compounds at risk for abuse, misuse, and diversion